Neural correlates of enhanced auditory sentence comprehension by binaural beat stimulation

Jeahong Kim, Hyun-Woong Kim, Jessica Kovar, & Yune Sang Lee
School of Behavioral and Brain Sciences, The University of Texas at Dallas; Callier Center for Communication Disorders, The University of Texas at Dallas

Background

- Beta and gamma oscillations are thought to reflect syntactic and semantic processing, respectively[1].
- Our previous behavioral study reported a positive effect of beta and gamma binaural beats (BB) on an auditory sentence comprehension task[2].
- Binaural beat - a de novo sound generated by the brain upon dichotic presentation of slightly mismatched tones[3] - have been shown to improve cognition[4] such as working memory[5] and attention[6].
- Neural mechanisms underlying enhanced auditory sentence comprehension by these binaural beats remain elusive.

Methods

BINAURAL BEAT STIMULATION

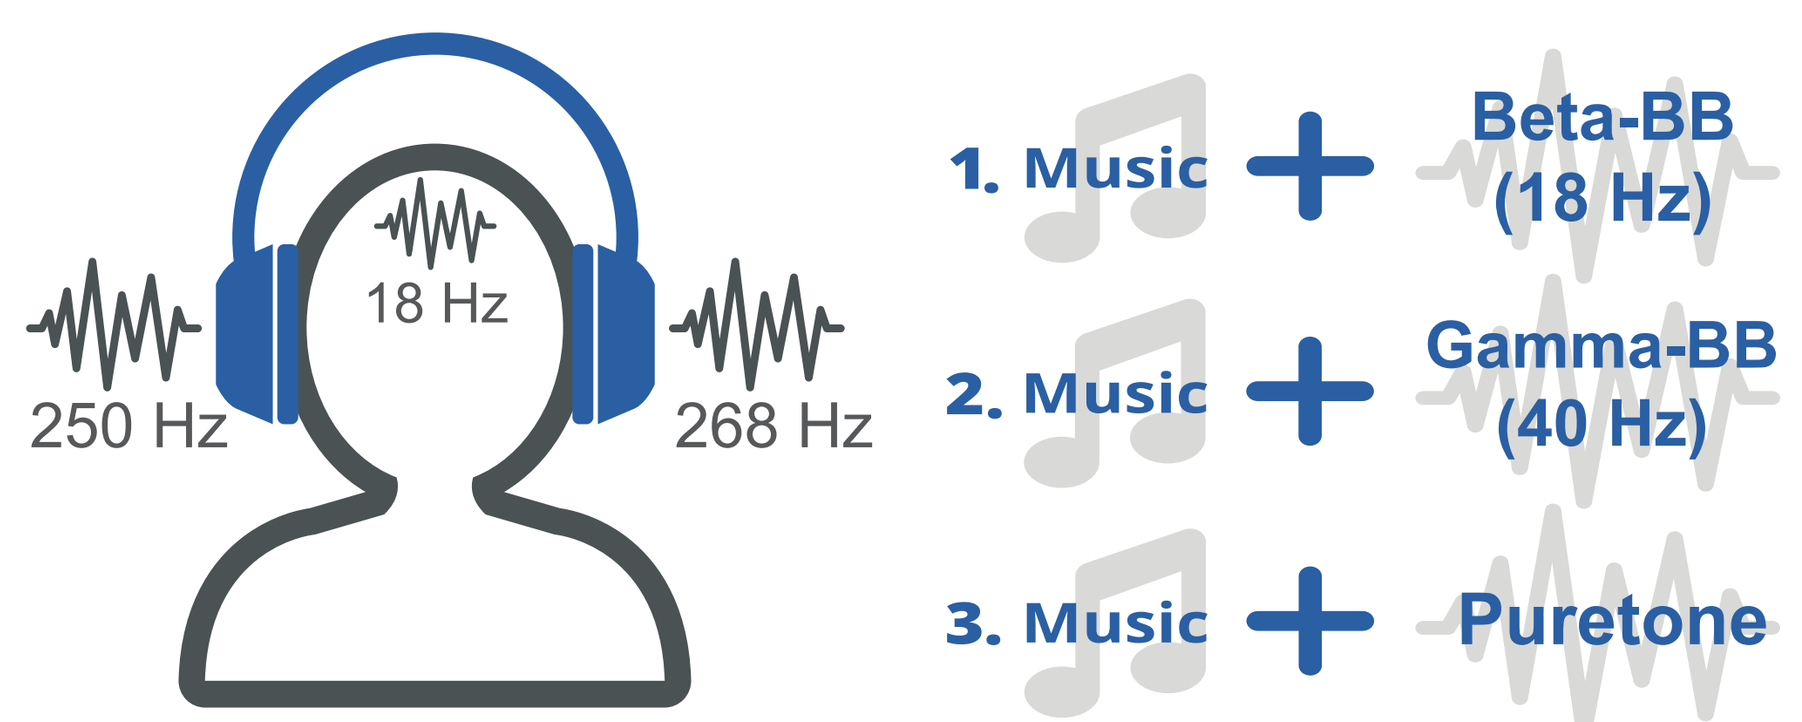
250 Hz
18 Hz
268 Hz
1. Music + Beta-BB (18 Hz)
2. Music + Gamma-BB (40 Hz)
3. Music + Puretone

PARTICIPANTS

60 UTD students
(mean age: 24.6 years, *SD*: 2.7)

Participants were randomly assigned to one of three auditory conditions (*N*=20 for each).

EXPERIMENTAL PROCEDURES

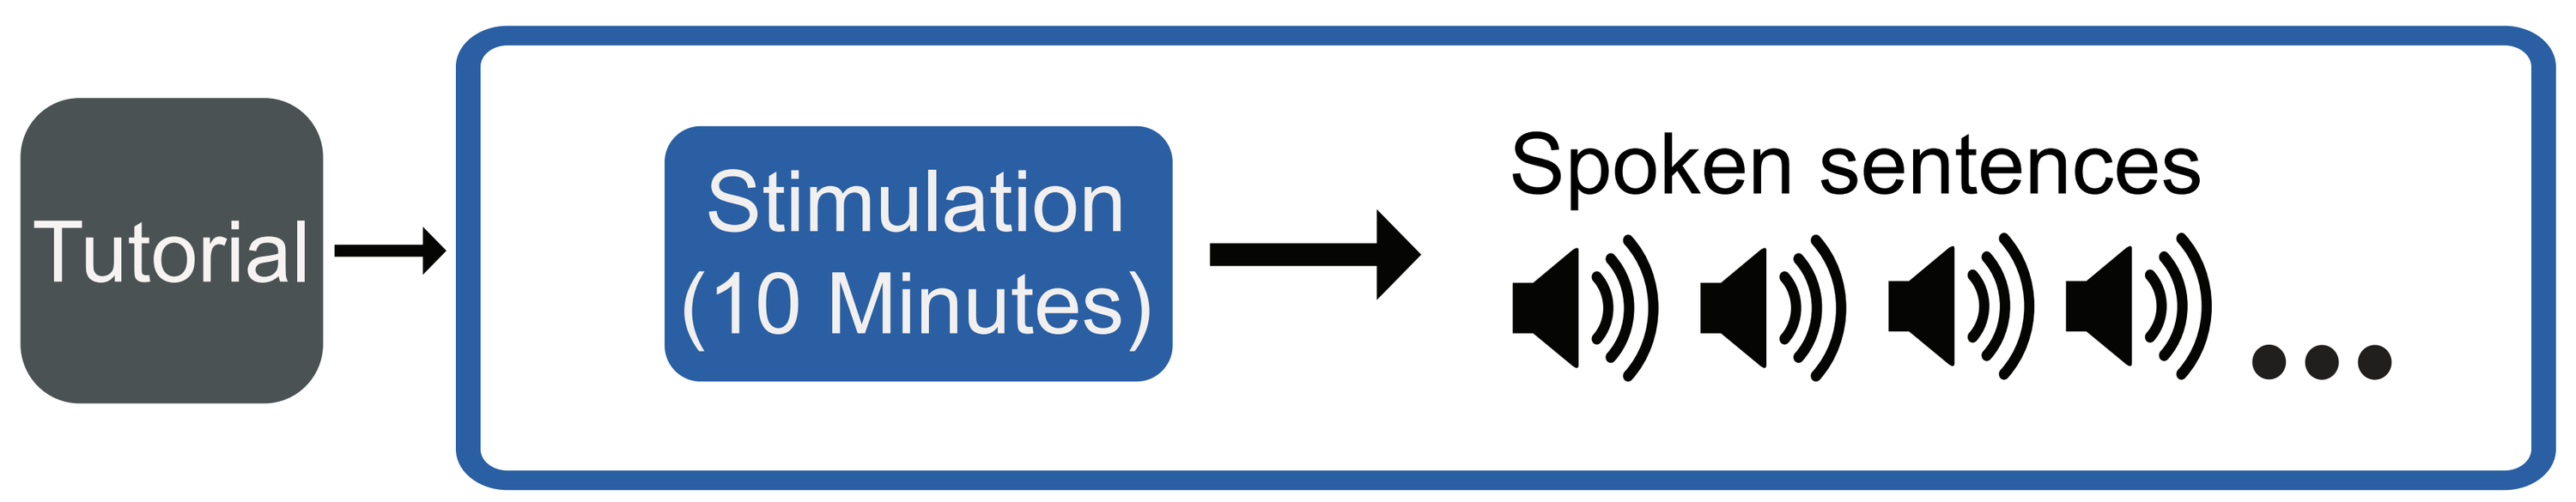
Tutorial
Stimulation (10 Minutes)
Spoken sentences

AUDITORY SENTENCE COMPREHENSION

Participants identified either gender of the *persons* or gender of the *agents* performing an **action**.

The two sentence types only differed in the order of the noun and the **verb** within the relative clause.

Sentence Type	Example	Answer
Subject relative (SR)	Boys that **join** aunts are lucky	Male
	Aunts that **join** boys are lucky	Female
Object relative (OR)	Boys that aunts **join** are lucky	Female
	Aunts that boys **join** are lucky	Male

Hypotheses

- **Behavioral task**: Beta and gamma binaural beats will enhance auditory sentence comprehension task performance, especially for sentences that are more difficult to process.
- **EEG dataset**: Beta and gamma binaural beats will entrain beta and gamma frequency ranges both in EEG-stimulation period and EEG-task period compared to the control tone. Due to their distinct functional roles in language processing, beta oscillations are known to modulate syntactic processing, while gamma oscillations modulate semantic processing[7].

Results

BEHAVIORAL

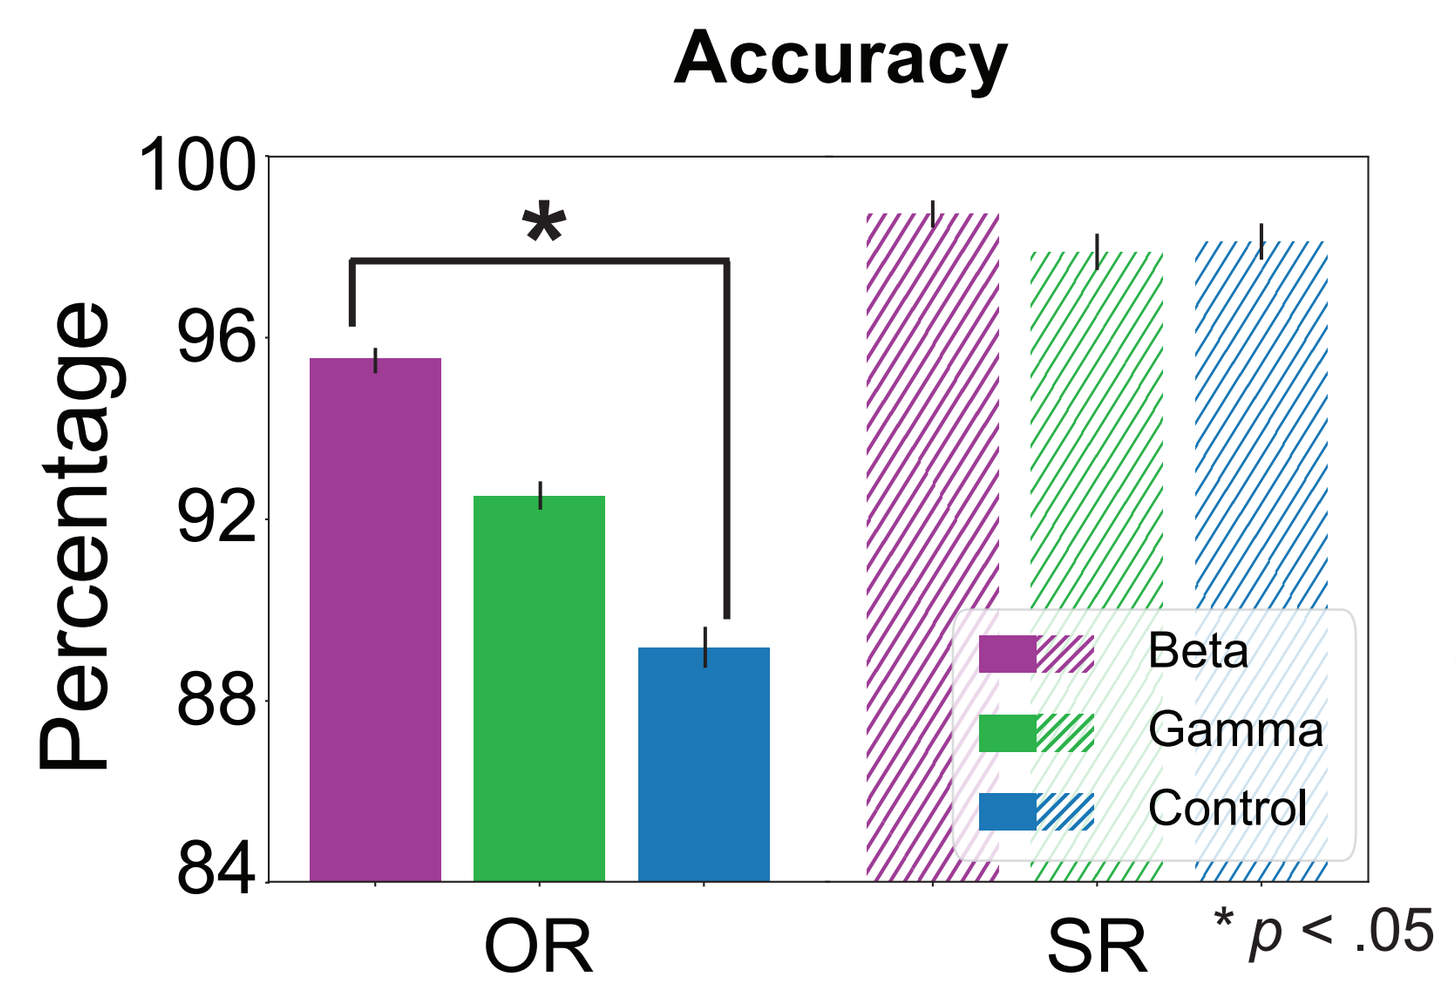
Accuracy
Percentage
100
96
92
88
84
*
Beta
Gamma
Control
OR
SR
* p < .05

Sentence type, $p < .001$
Auditory condition, $p = .050$
Interaction, $p = .064$

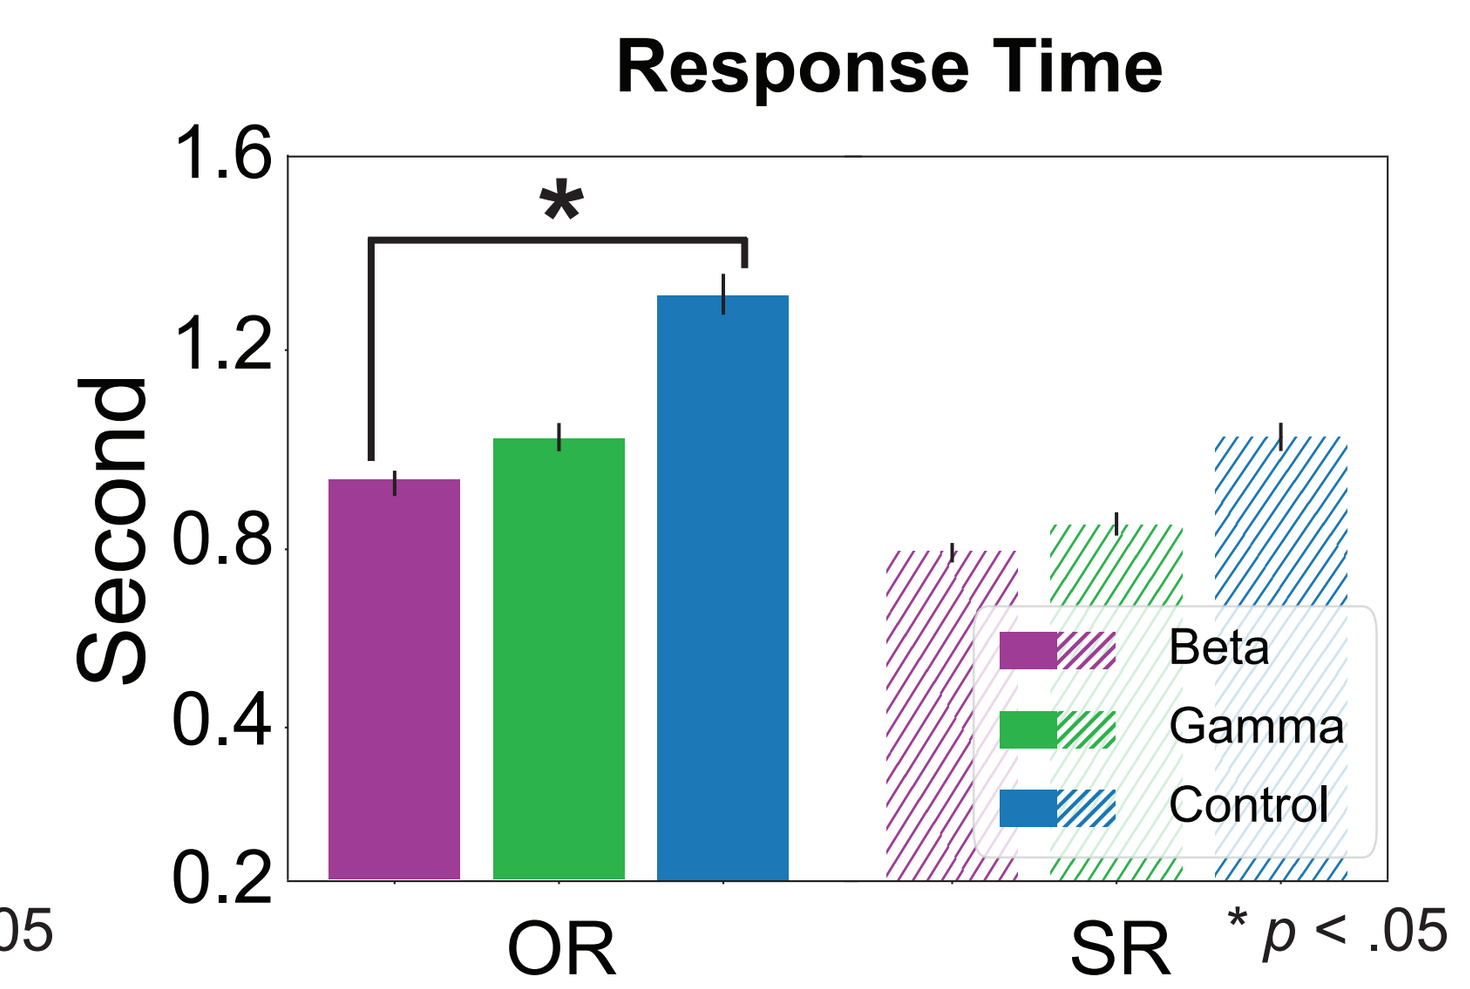
Response Time
Second
1.6
1.2
0.8
0.4
0.2
*
Beta
Gamma
Control
OR
SR
* p < .05

Sentence type, $p < .001$
Auditory condition, $p = .024$
Interaction, $p < .001$

EEG: STIMULATION PERIOD

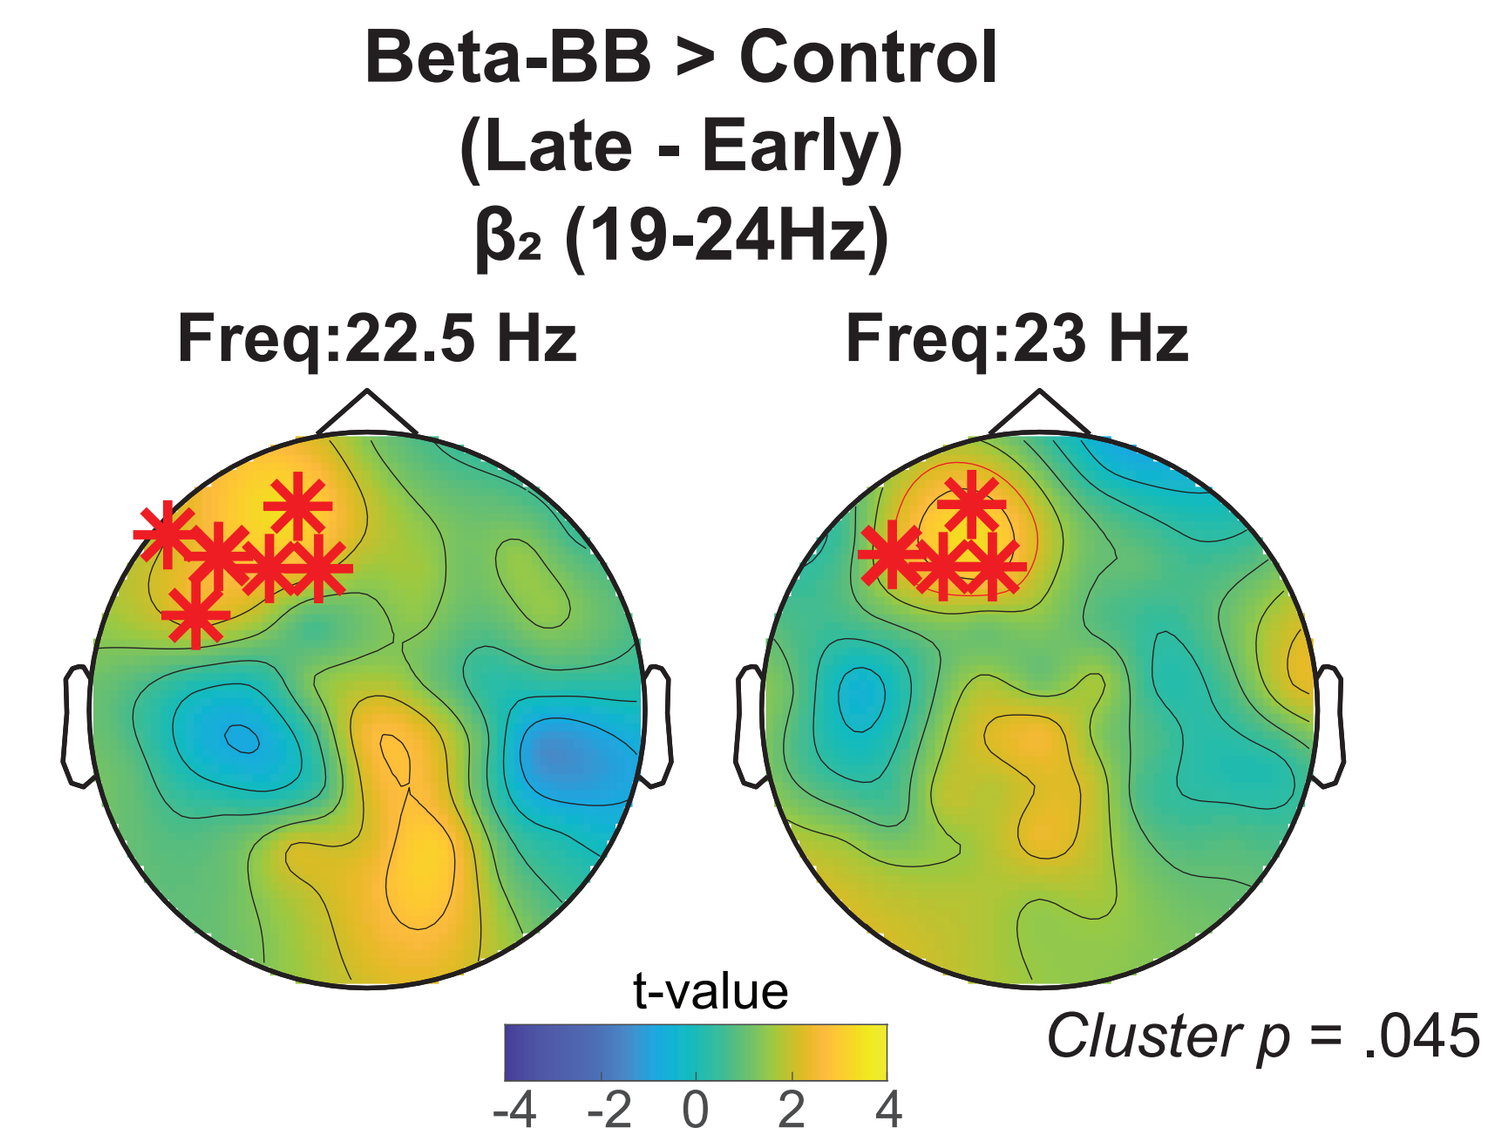
Beta-BB > Control
(Late - Early)
β2 (19-24Hz)
Freq:22.5 Hz
Freq:23 Hz
t-value
-4 -2 0 2 4
Cluster p = .045

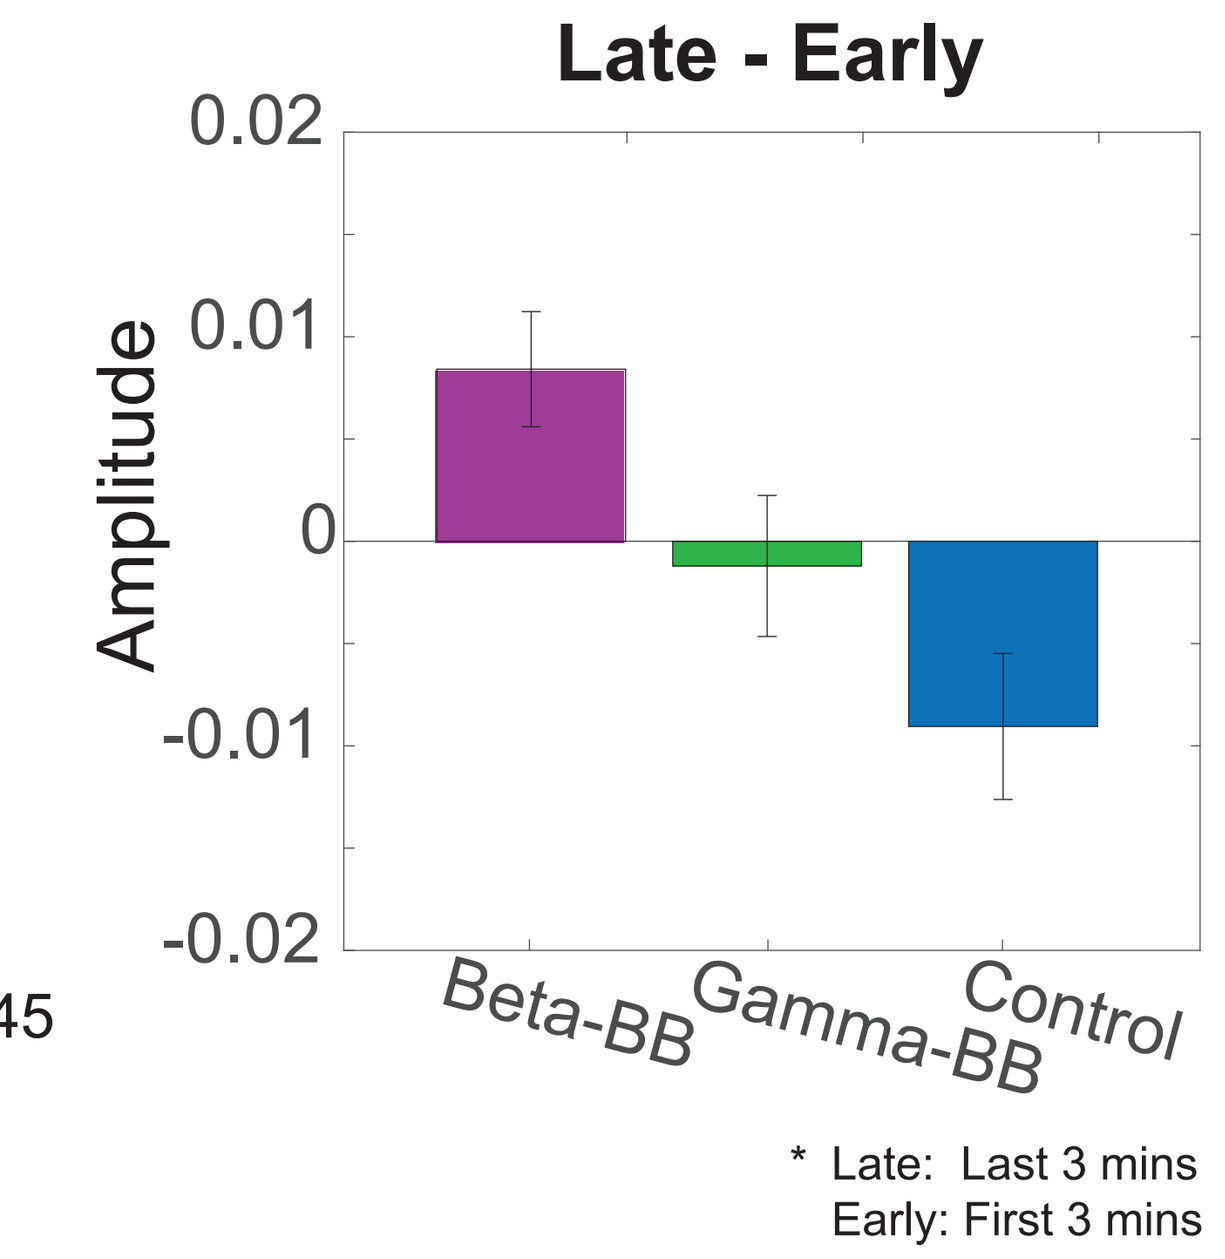
Late - Early
Amplitude
0.02
0.01
0
-0.01
-0.02
Beta-BB
Gamma-BB
Control
* Late: Last 3 mins
Early: First 3 mins

EEG: LANGUAGE TASK PERIOD

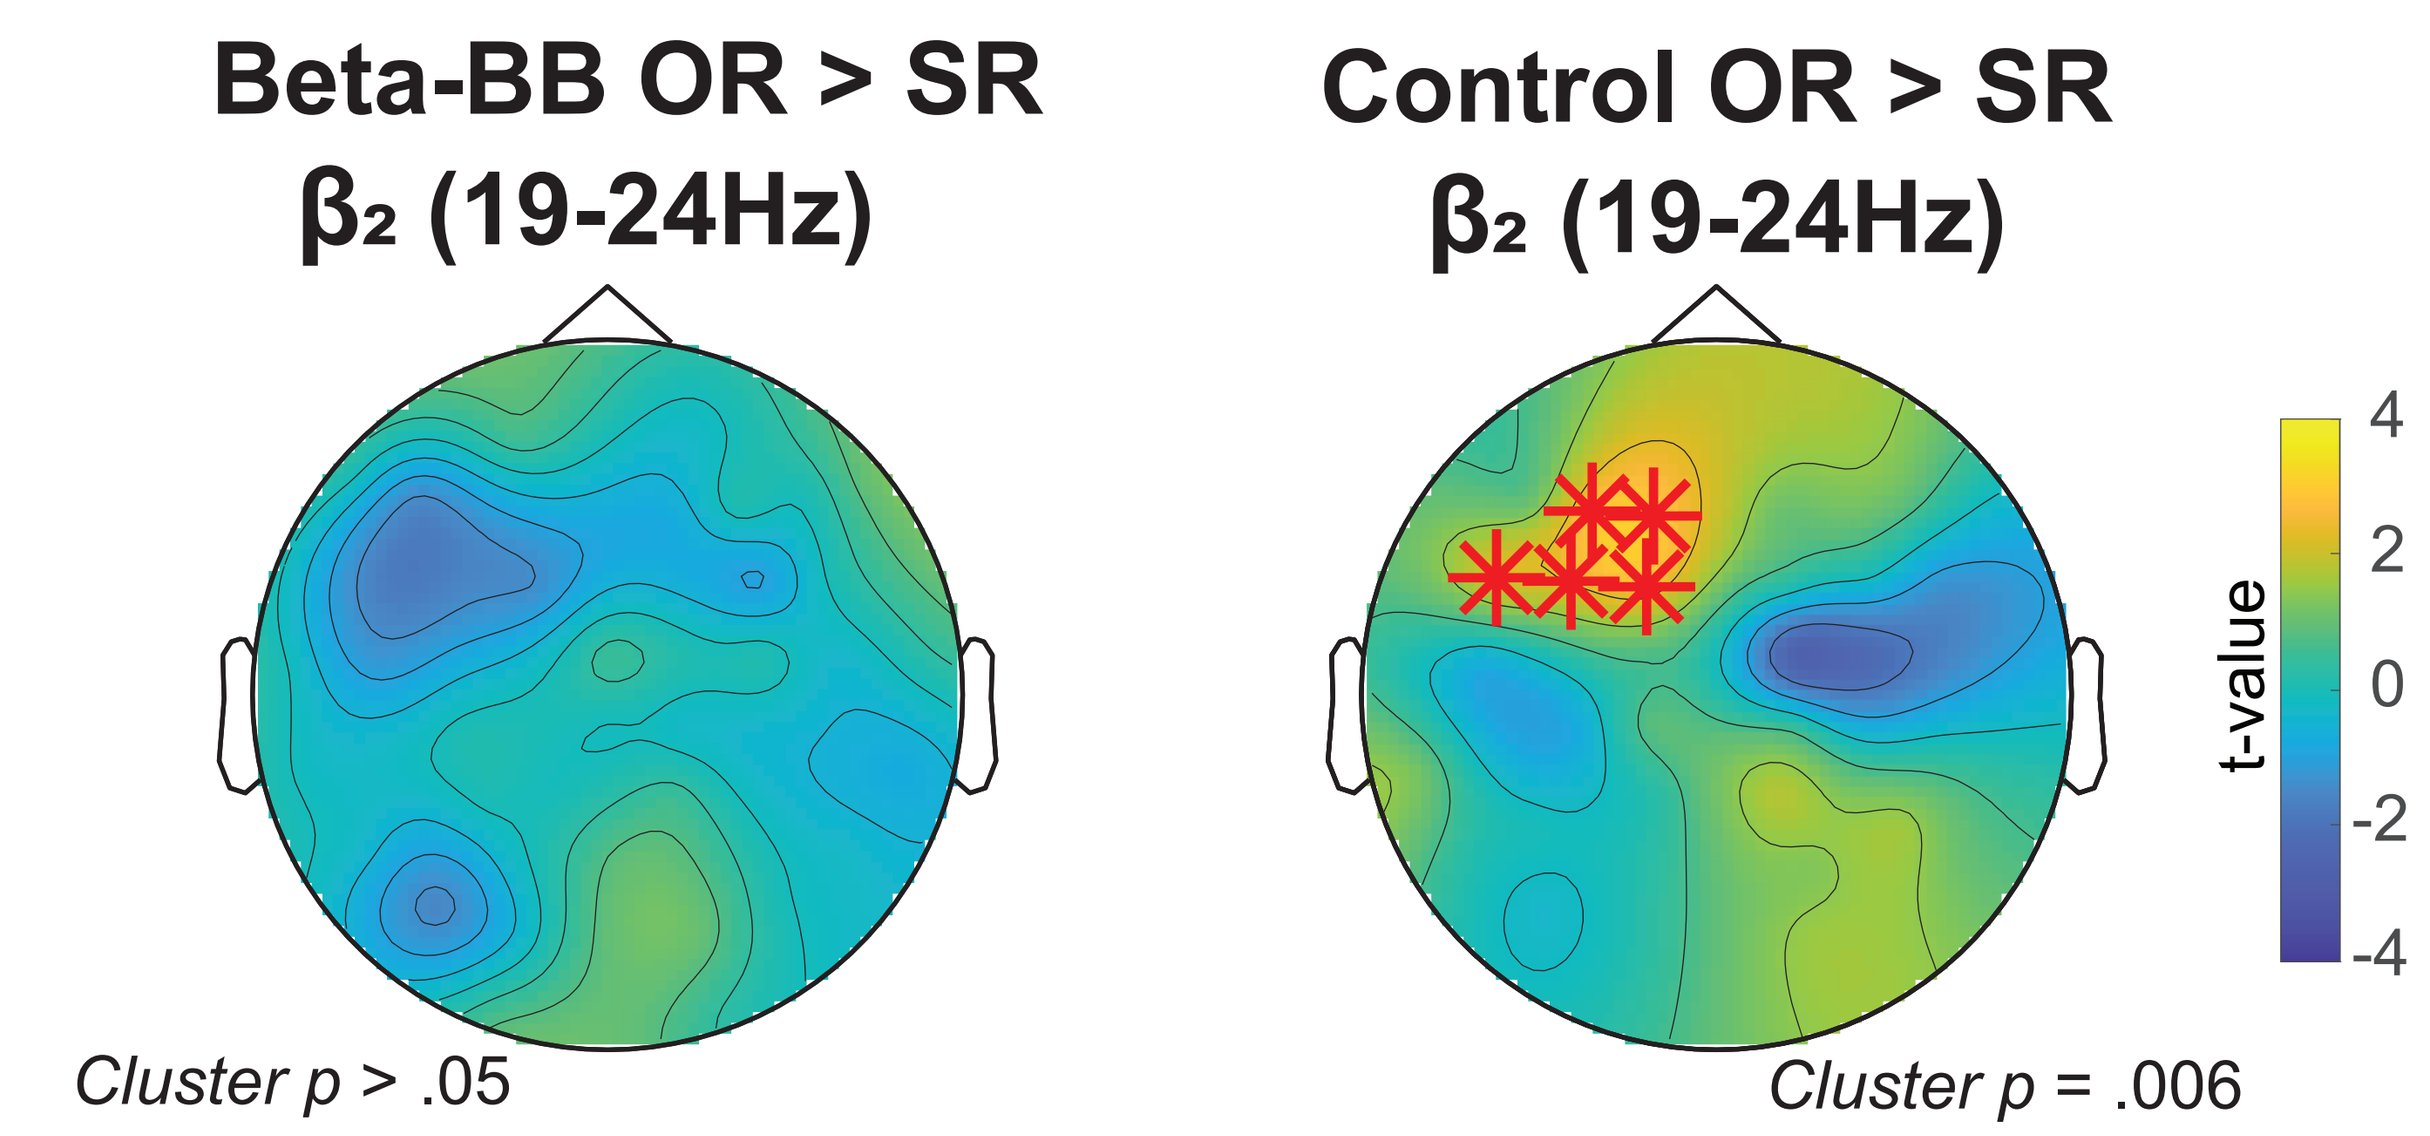
Beta-BB OR > SR
β2 (19-24Hz)
Control OR > SR
β2 (19-24Hz)
t-value
4
2
0
-2
-4
Cluster p > .05
Cluster p = .006

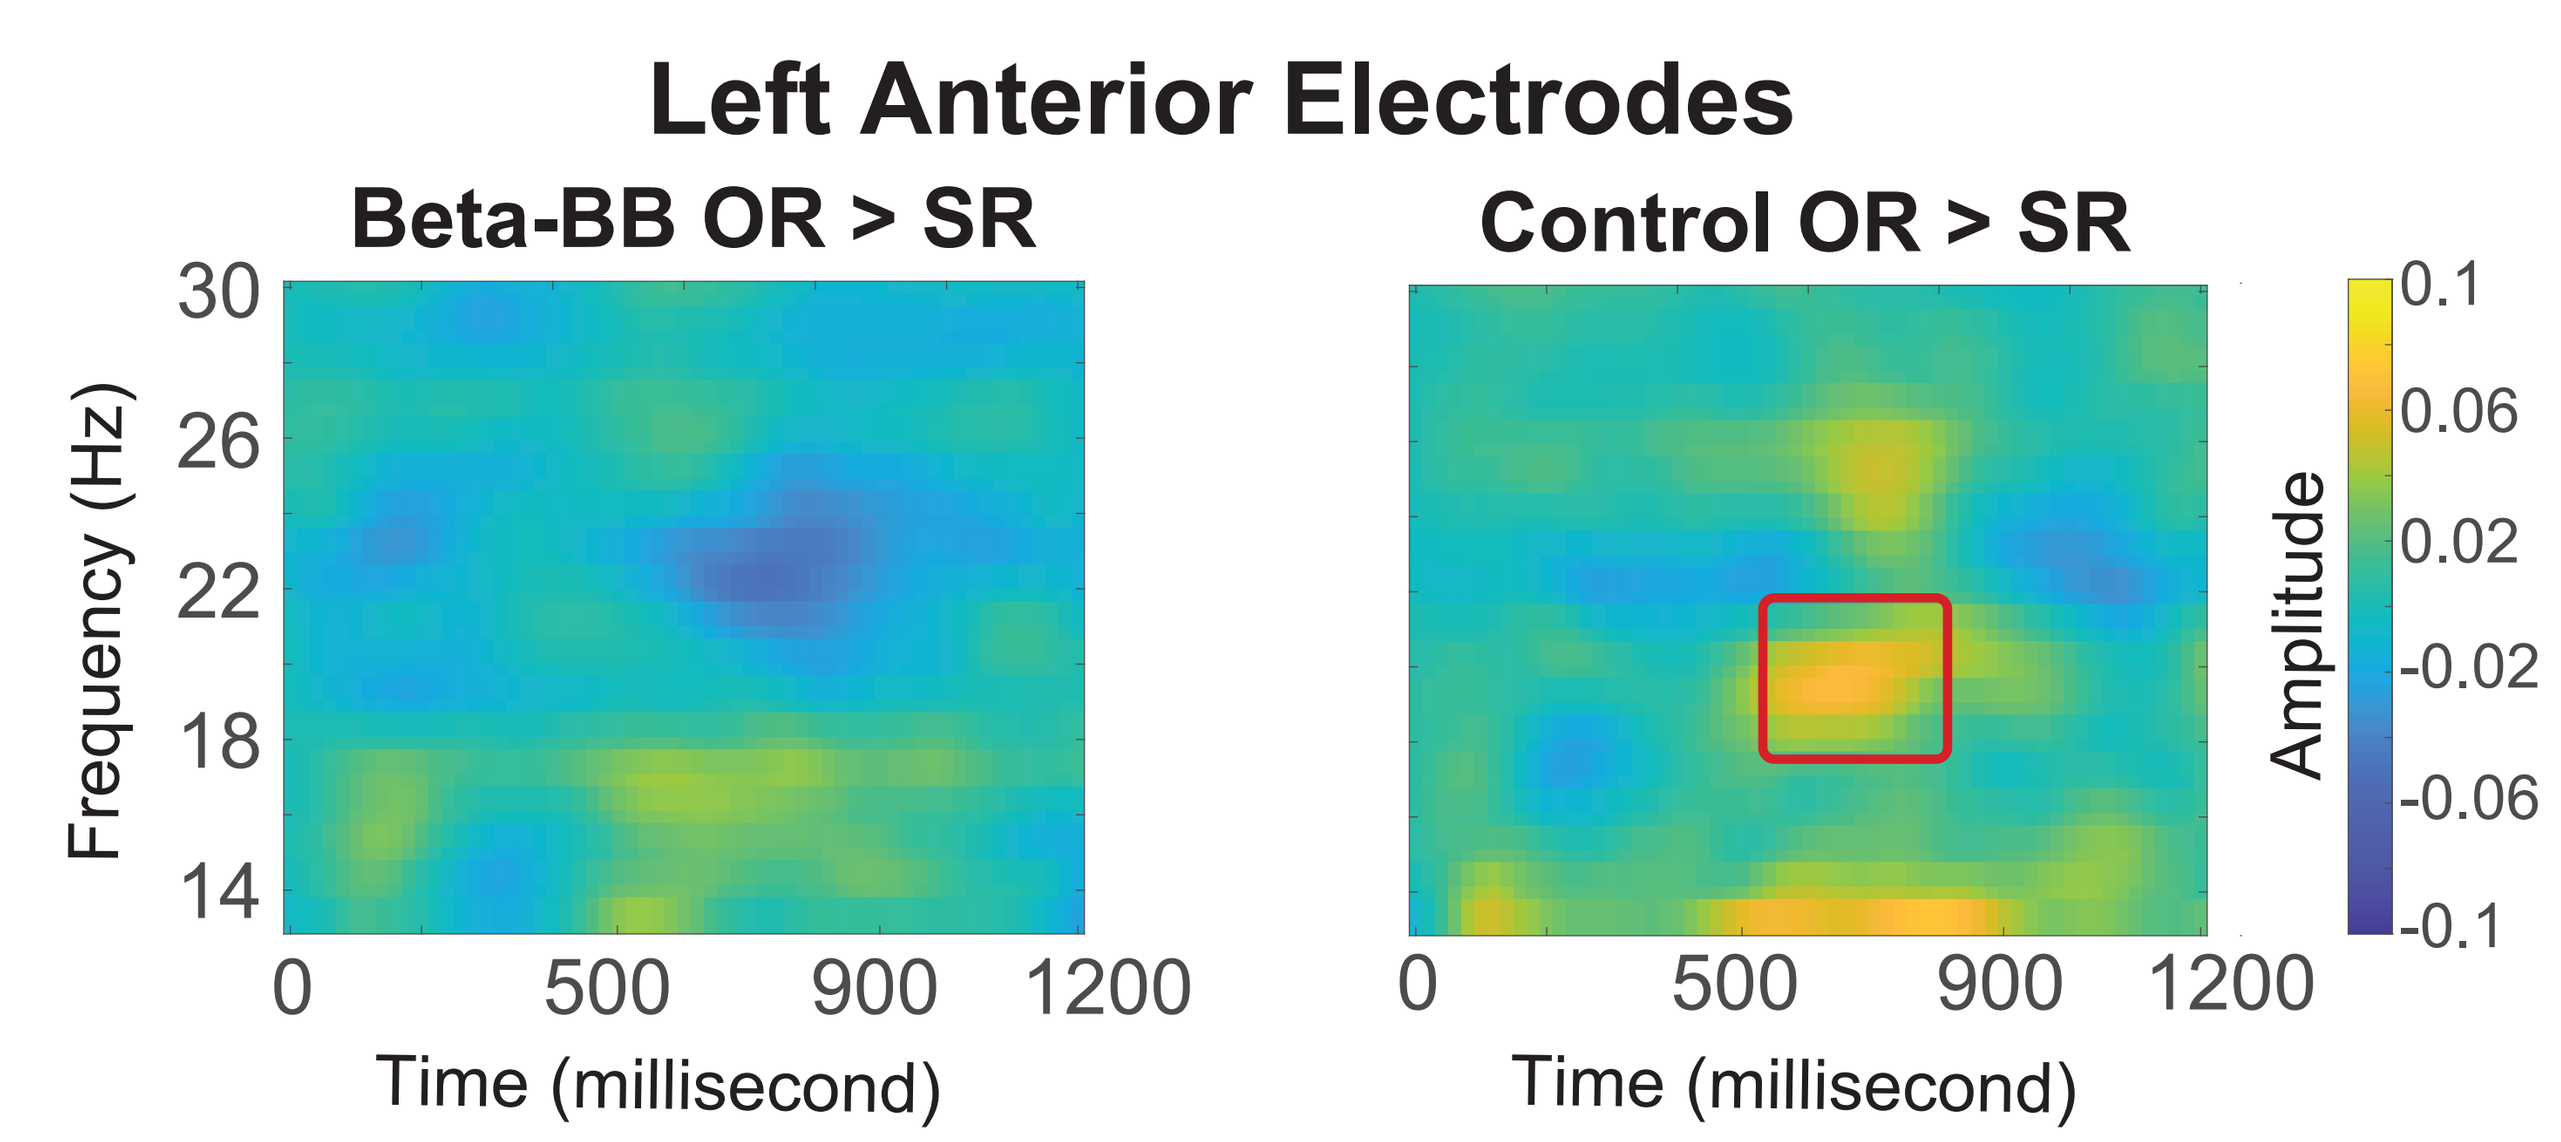
Left Anterior Electrodes
Beta-BB OR > SR
Control OR > SR
Frequency (Hz)
30
26
22
18
14
0
500
900
1200
Time (millisecond)
Amplitude
0.1
0.06
0.02
-0.02
-0.06
-0.1

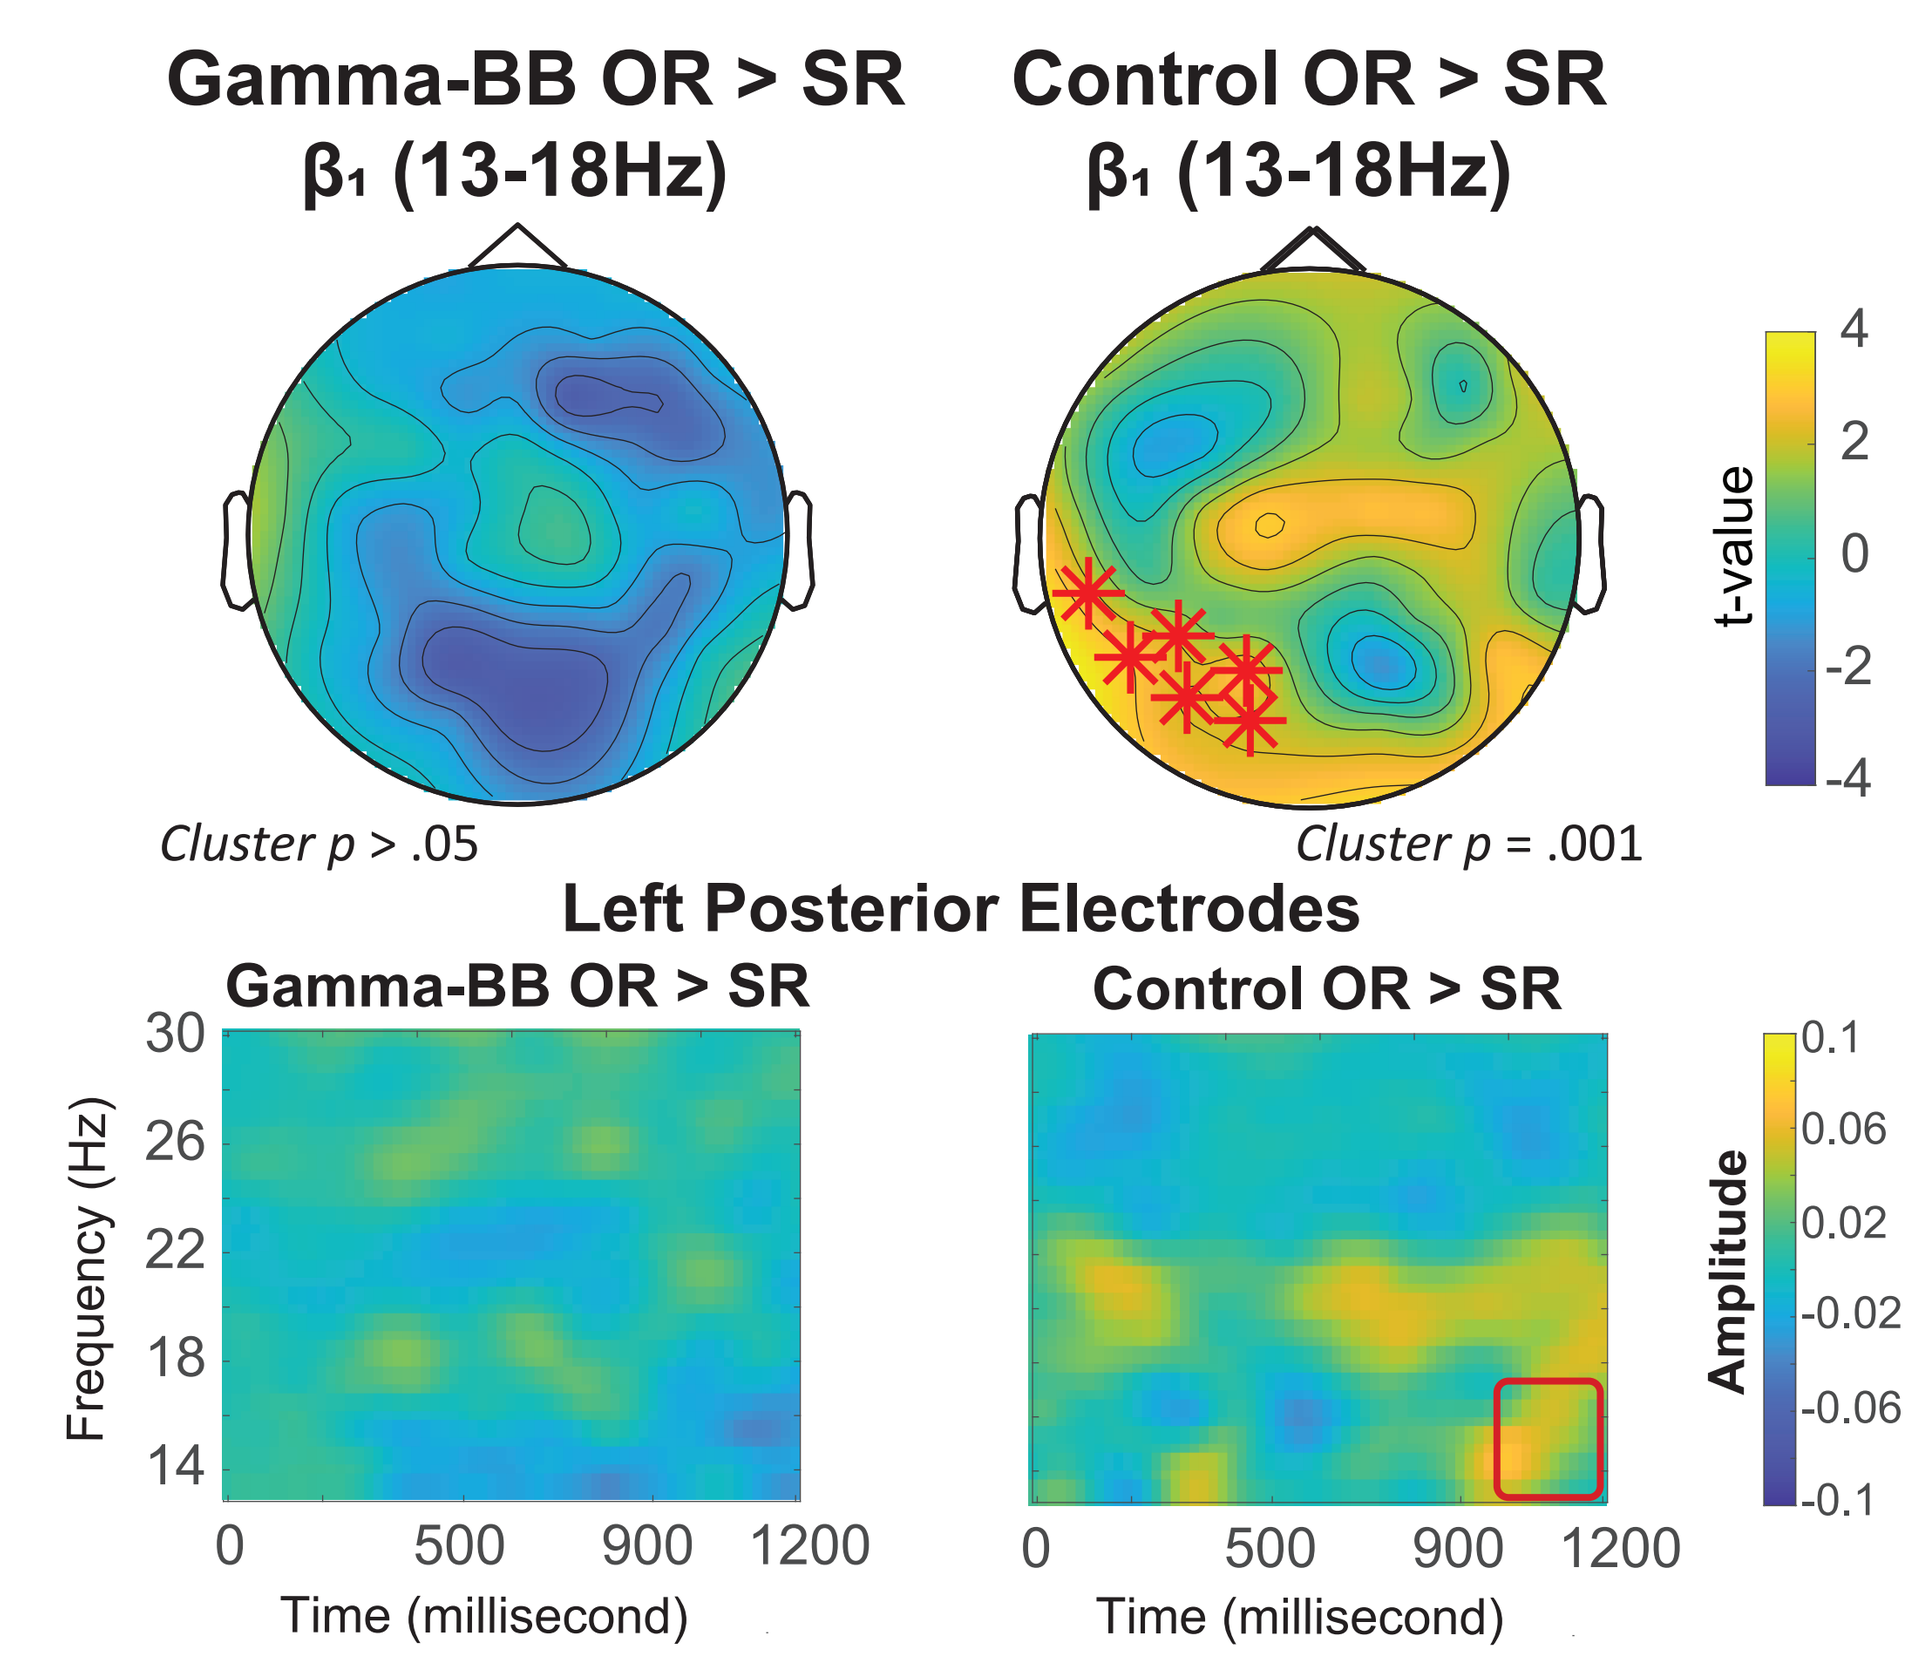
Gamma-BB OR > SR
β1 (13-18Hz)
Control OR > SR
β1 (13-18Hz)
t-value
4
2
0
-2
-4
Cluster p > .05
Cluster p = .001
Left Posterior Electrodes
Gamma-BB OR > SR
Control OR > SR
Frequency (Hz)
30
26
22
18
14
0
500
900
1200
Time (millisecond)
Amplitude
0.1
0.06
0.02
-0.02
-0.06
-0.1

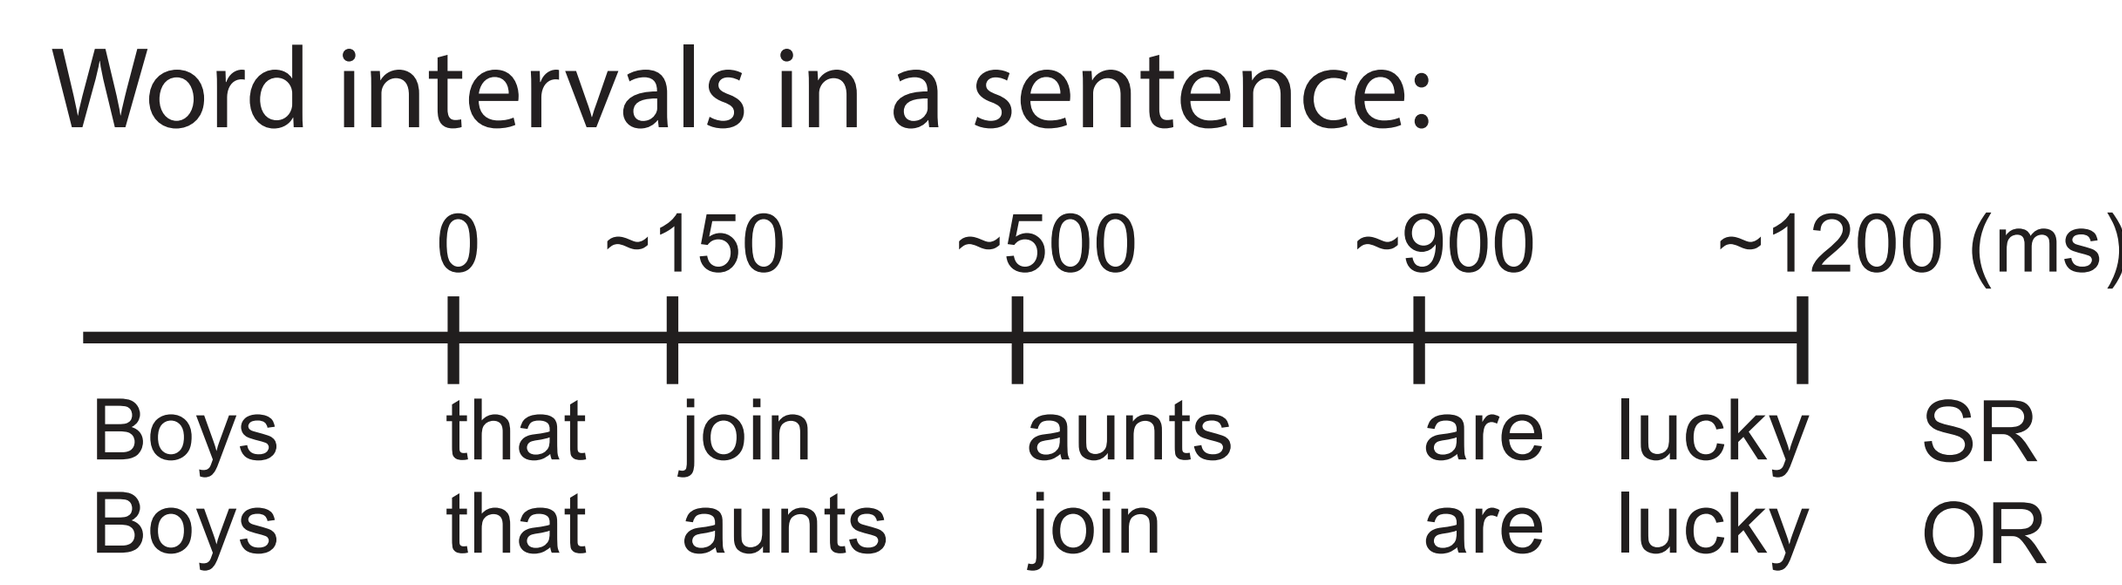
Word intervals in a sentence:
0
~150
~500
~900
~1200 (ms)
Boys that join aunts are lucky SR
Boys that aunts join are lucky OR

CORRELATIONS

Stimulation EEG & Task

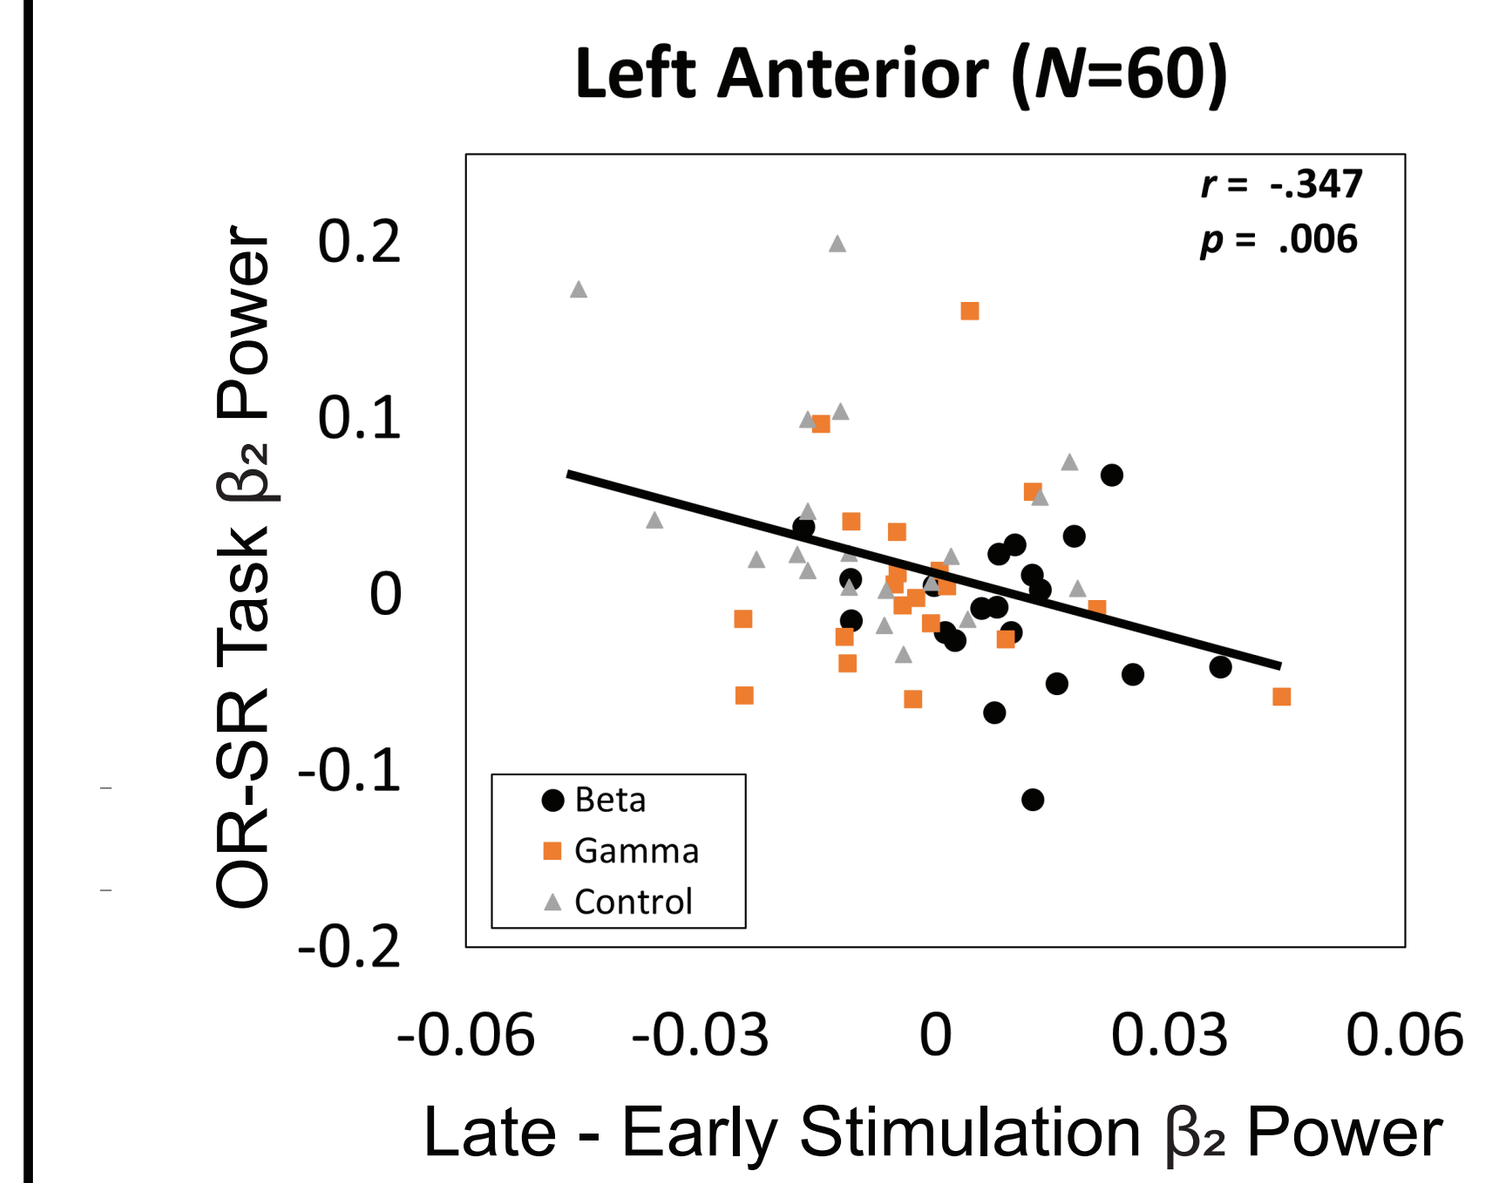
Left Anterior (N=60)
r = -.347
p = .006
OR-SR Task β2 Power
0.2
0.1
0
-0.1
-0.2
Beta
Gamma
Control
-0.06
-0.03
0
0.03
0.06
Late - Early Stimulation β2 Power

Task EEG & Behavioral

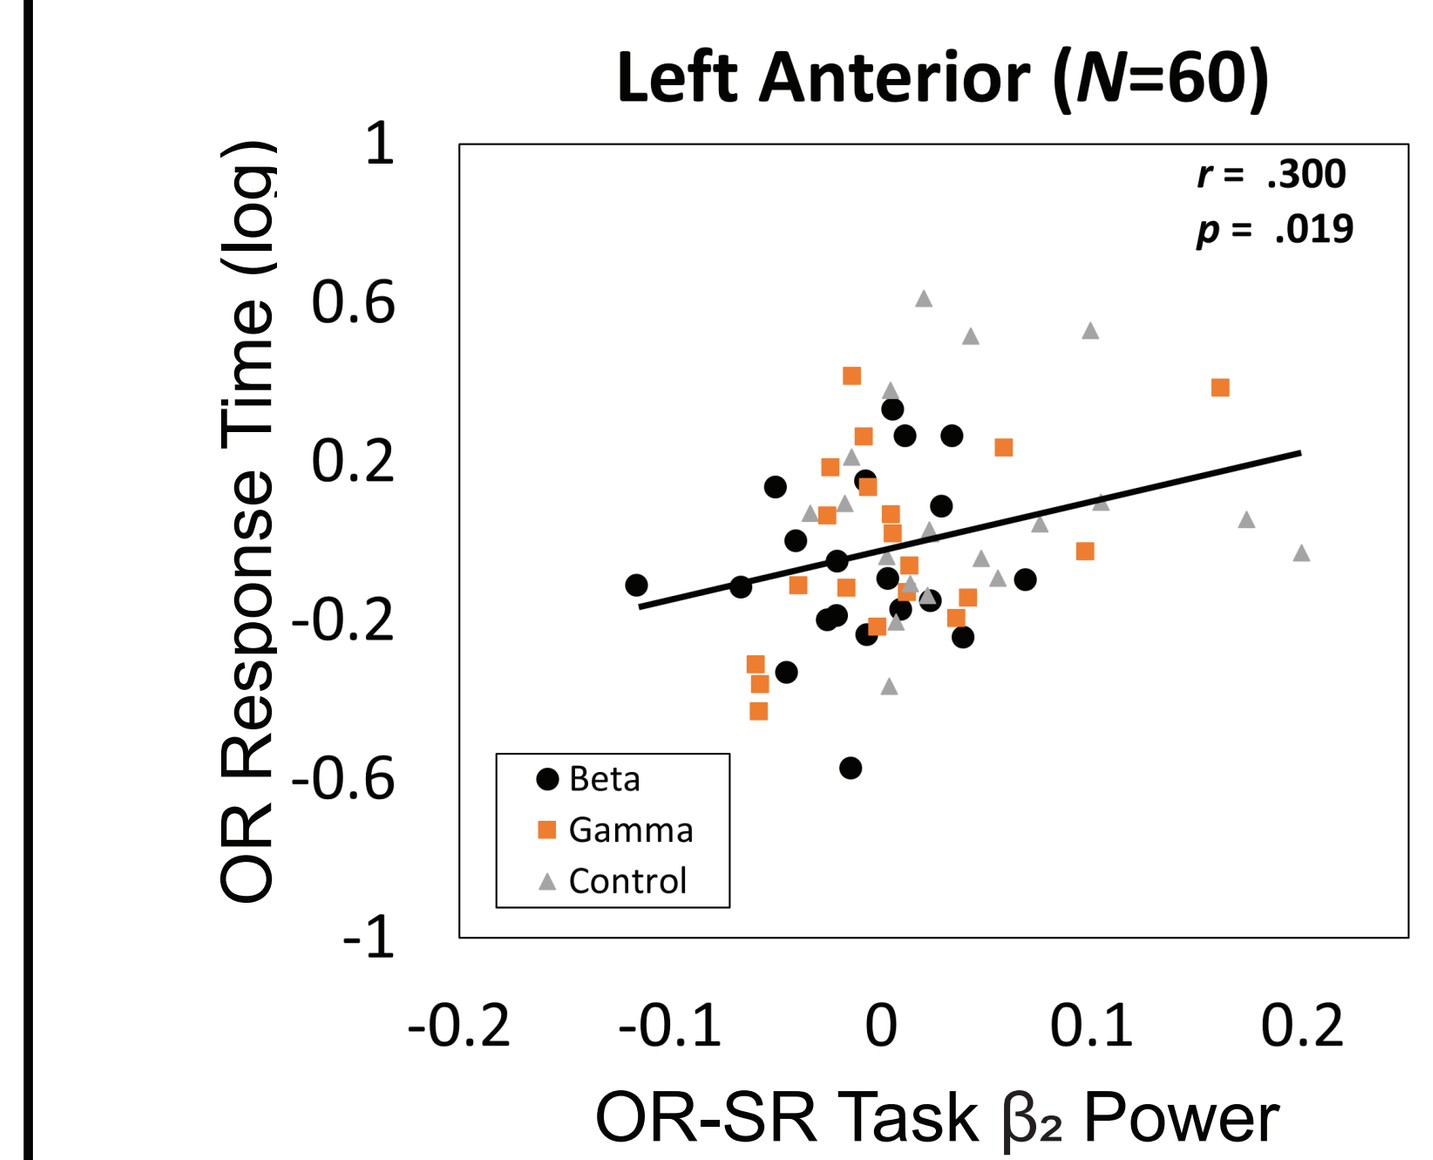
Left Anterior (N=60)
r = .300
p = .019
OR Response Time (log)
1
0.6
0.2
-0.2
-0.6
-1
Beta
Gamma
Control
-0.2
-0.1
0
0.1
0.2
OR-SR Task β2 Power

Stimulation EEG & Behavioral

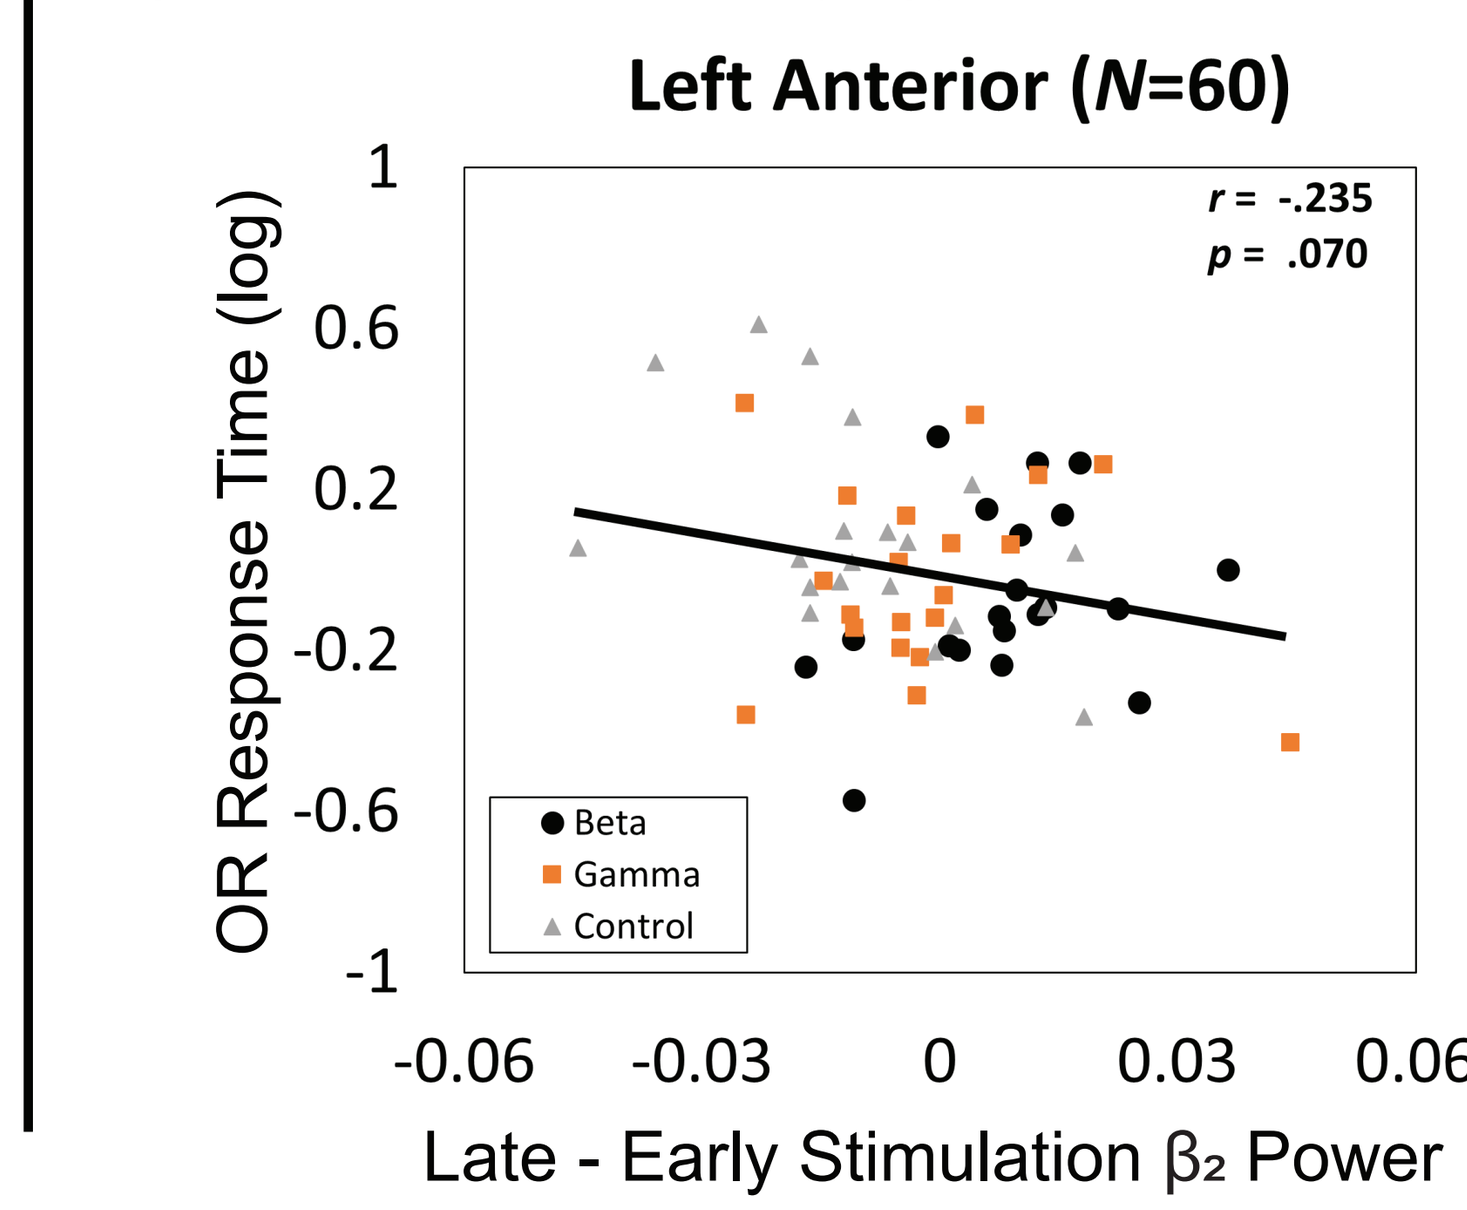
Left Anterior (N=60)
r = -.235
p = .070
OR Response Time (log)
1
0.6
0.2
-0.2
-0.6
-1
Beta
Gamma
Control
-0.06
-0.03
0
0.03
0.06
Late - Early Stimulation β2 Power

Discussion

- We replicated our previous behavioral findings on the positive impact of beta-BB on sentence comprehension[2], even with a smaller sample size.
- The behavioral enhancement was manifested in both increased accuracy and reduced response time.
- However, we were unable to replicate the gamma-BB effect, possibly due to the smaller sample size.
- During beta-BB stimulation, increased power at β_2 frequency (19-24 Hz) was observed in the left anterior region, which was also associated with β_2 power modulation during the language task.
- Reduced β_2 power difference between OR and SR was correlated with improved task performance, suggesting that beta-BB reduced task burden on OR processing.

REFERENCES

1. Prystauka & Lewis (2019). Language and Linguistics Compass, 13(9).
2. Kim et al. (2023). Psychological Research, 1-10.
3. Oster (1973). Scientific American. 229(4), 94-102.
4. Ala et al. (2018). Biomedical Signal Processing and Control, 42, 242-252.
5. Ortiz et al. (2008). Actas Espanolas de Psiquiatria, 36(6), 307–313.
6. Garcia-Argibay et al. (2019). Psychological Research 83, 357–372.
7. Bastiaansen et al., (2010). Journal of Cognitve Neuroscience, 22(7), 1333-1347.

ACKNOWLEDGEMENTS

We thank Digisonic and the Neuroscience Innovation Foundation for supporting this research.

Email: jeahong.kim@utdallas.edu